A Phase II/III Randomized, Open-Label Study of Toca 511, a Retroviral Replicating Vector, Combined With Toca FC with Temozolomide and Radiation followed by adjuvant Temozolomide and Toca FC compared to Temozolomide and Radiation followed by adjuvant Temozolomide in Patients with Newly Diagnosed Glioblastoma

NRG ONCOLOGY

Advancing Research. Improving Lives.™

Manmeet S. Ahluwalia, MD, Stephanie Pugh, PhD, Benjamin M. Ellingson, PhD, Rupesh Kotecha, MD, Timothy F. Cloughesy, MD, Michael A. Vogelbaum, MD, PhD, Yunfeng Cui, PhD, Ken Aldape, MD, Terri S. Armstrong, PhD, Minesh P. Mehta, MD

ABSTRACT

The standard of care (SOC) in newly diagnosed glioblastoma (ndGBM) includes resection and chemoradiotherapy. With a median overall survival (OS) of only 16-18 months for well-selected patients in clinical trials, better therapeutic options are needed. Toca 511 (vocimagene amiretrorepvec) is a retroviral replicating vector encoding a codon optimized, heat stabilized cytosine deaminase that converts Toca FC (extended-release 5-fluorocytosine, 5-FC) into 5-fluorouracil. Preclinical evidence demonstrates that Toca 511 & 5-FC kill cancer cells and immunosuppressive myeloid cells in the tumor microenvironment, leading to durable antitumor immune responses. Three phase (P) 1 studies in addition to a randomized Phase 3 trial (Toca 5) in a total of 329 patients with recurrent high grade glioma have demonstrated an acceptable safety profile and encouraging signals of efficacy.

NRG-BN006 is a randomized P 2/3 trial of Toca 511 & Toca FC with SOC versus SOC for patients with ndGBM. Optune use is allowed on the SOC arm, but not on the experimental arm. Patients will be stratified by age and KPS score for 1:1 randomization. The primary endpoint is progression-free survival for P2 and OS for P3. The secondary endpoints include objective response rate in patients with measurable disease and safety. Key inclusion criteria include presumptive diagnosis of glioblastoma with anticipated 80% resection, unifocal tumor, and KPS≥70. Immune monitoring and molecular profiling will be performed. P2 has 90% power to detect a hazard ratio (HR)=0.67 in 250 ndGBM patients. P3 has 85% power to detect a HR=0.75 in 720 ndGBM patients. Since patients are enrolled prior to surgery and confirmatory diagnosis of GBM, approximately 900 patients will be enrolled, and two interim analyses are planned for OS. In addition, two interim safety analyses will be conducted for the experimental treatment, with the first 15 and 30 eligible and analyzable patients randomized to the experimental arm. NRG-BN006 is anticipated to start enrollment in Q4 2019. Supported by grants U10CA180868, U10CA180822 from NCI and Tocagen.

BACKGROUND

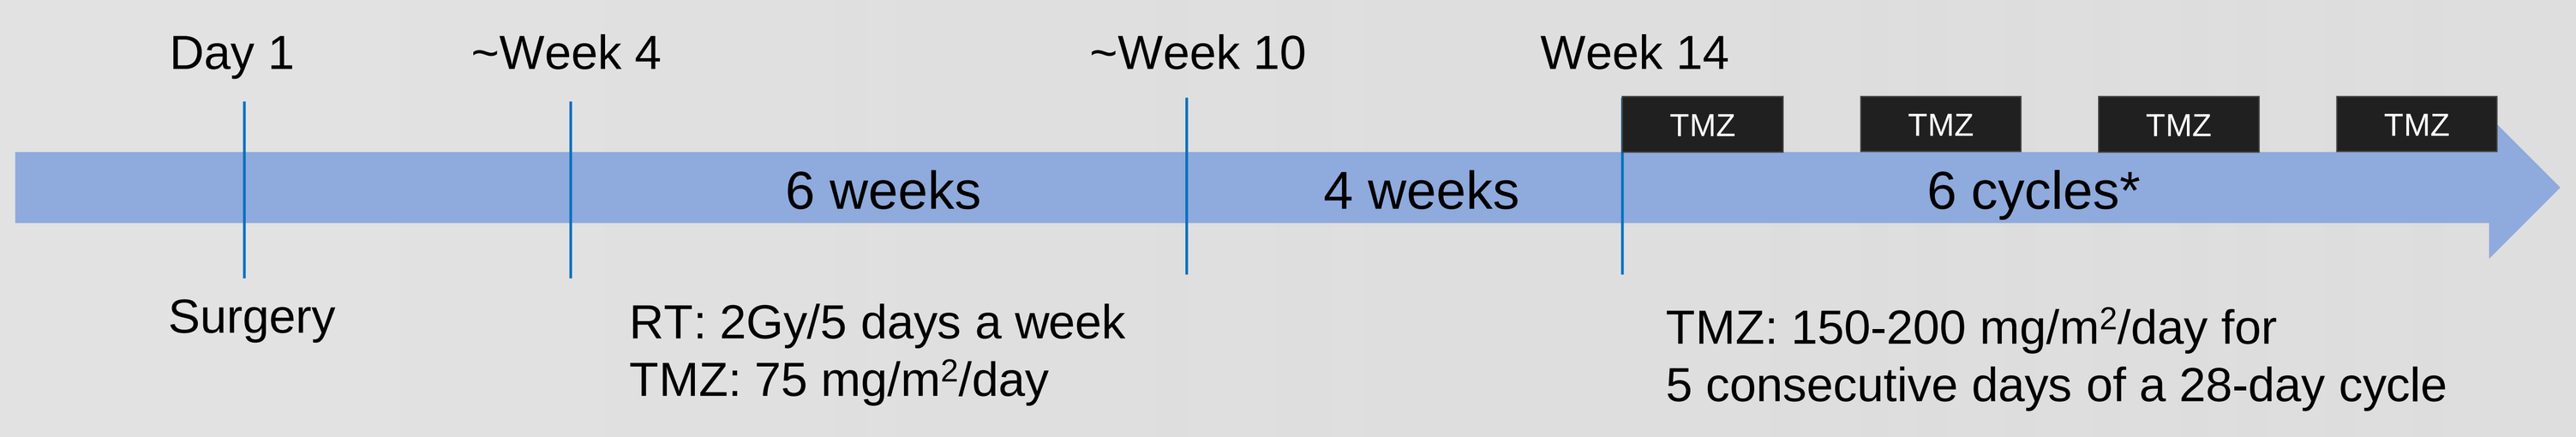
Day 1
~Week 4
~Week 10
Week 14
TMZ
TMZ
TMZ
TMZ
6 weeks
4 weeks
6 cycles*
Surgery
RT: 2Gy/5 days a week
TMZ: 75 mg/m2/day
TMZ: 150-200 mg/m2/day for
5 consecutive days of a 28-day cycle

Figure 1: Standard of Care Treatment for Patients with Newly Diagnosed Glioblastoma

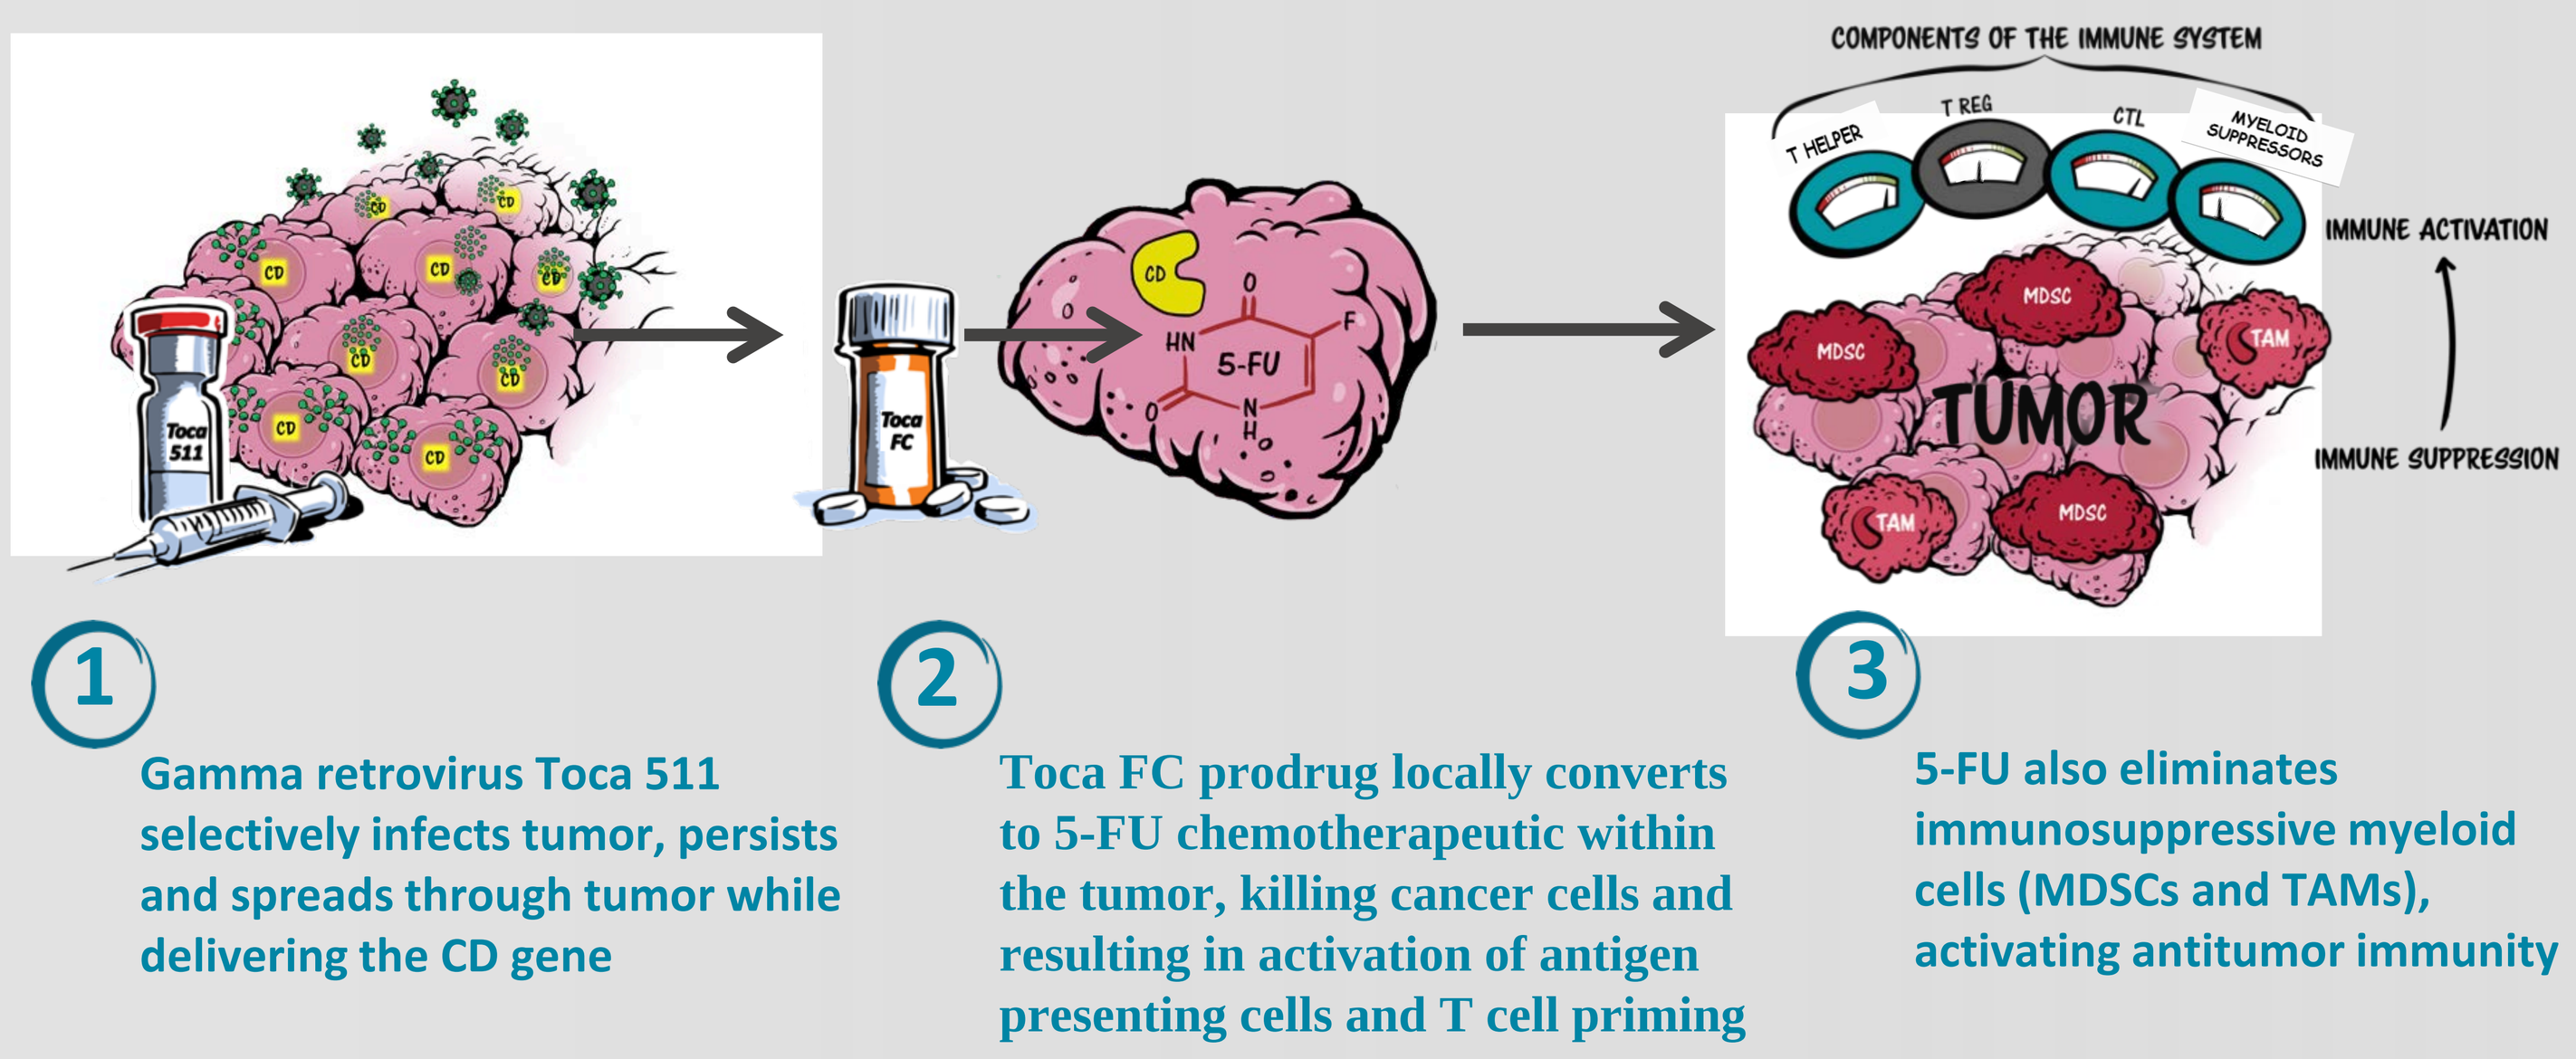
COMPONENTS OF THE IMMUNE SYSTEM
T HELPER
T REG
CTL
MYELOID SUPPRESSORS
IMMUNE ACTIVATION
IMMUNE SUPPRESSION
TUMOR
MDSC
TAM
5-FU
Toca 511
Toca FC
1
Gamma retrovirus Toca 511 selectively infects tumor, persists and spreads through tumor while delivering the CD gene
2
Toca FC prodrug locally converts to 5-FU chemotherapeutic within the tumor, killing cancer cells and resulting in activation of antigen presenting cells and T cell priming
3
5-FU also eliminates immunosuppressive myeloid cells (MDSCs and TAMs), activating antitumor immunity

Figure 2: Proposed Mechanism of Action for Toca 511 & Toca FC

- **Toca 511**
 - Amphotropic gammaretrovirus-based retroviral replicating vector (RRV) containing a modified yeast CD transgene
 - Designed to selectively spread through cancer cells and stably deliver an optimized CD gene whose protein product converts the prodrug Toca FC into 5-fluorouracil (5-FU)
- **Toca FC**
 - an investigational extended-release version of flucytosine; 5-fluorocytosine, 5-FC
 - Flucytosine is an oral antifungal drug that crosses the blood brain barrier and is approved to treat patients with fungal infections of the central nervous system.

OBJECTIVES

Primary Objectives:

- Phase II– To compare the progression-free survival (PFS) of patients with newly diagnosed glioblastoma treated with Toca 511 at the time of tumor resection followed by Toca FC in combination with standard of care (SOC) treatment to patients with newly diagnosed glioblastoma treated with SOC after tumor resection
- Phase III – To compare the overall survival (OS) from time of randomization of all patients with newly diagnosed glioblastoma treated with Toca 511 at the time of tumor resection followed by Toca FC in combination with SOC treatment to patients with newly diagnosed glioblastoma treated with SOC after tumor resection.

Secondary Objectives:

- To evaluate the safety of each arm as administered in this study
- To compare the OS between arms (only for phase II part of the study)
- To compare the PFS between arms using mRANO (only for phase III part of the study)
- To evaluate the objective response rate (ORR) in patients with measurable disease
- To evaluate the effect of IDH mutation status on survival outcomes between arms
- To evaluate the PFS and OS in those with gross total or near gross total resection (defined by less than 10 mm x 10 mm of enhancing disease) compared to those with ≥ than 10 mm x 10 mm of enhancing disease
- To evaluate the effect of MGMT methylation status on survival outcomes between arms

ELIGIBILITY

Key Eligibility Criteria:

- Presumptive diagnosis of glioblastoma based on MRI imaging within 14 days prior to registration.
- Patients who have undergone biopsy with diagnosis of glioblastoma and who have never received any chemotherapy and/or radiation and are candidates for ≥80% resection of enhancing region.
- The tumor must be unifocal, confined to the supratentorial compartment and based on the pre-operative evaluation the patient is a candidate for ≥80% resection of enhancing region.
- Measurable disease preoperatively, defined as at least 1 contrast-enhancing lesion, with 2 perpendicular measurements of at least 1 cm, as per RANO criteria.

Key Ineligibility Criteria:

- A contrast-enhancing brain tumor on MRI that is any of the following:
 - Multi-focal (defined as 2 separate areas of contrast enhancement measuring at least 10 mm in 2 planes that are not contiguous on either fluid-attenuated inversion recovery (FLAIR) or T2 hyperintensity);
 - Associated with either diffuse subependymal or leptomeningeal dissemination; or
 - > 5 cm in any dimension
- Active infection (excluding skin or toenail infections) requiring systemic antibiotic, antifungal or antiviral therapy within 28 days prior to registration
- Bleeding diathesis, or must take anticoagulants, or antiplatelet agents, including nonsteroidal anti-inflammatory drugs (NSAIDs), at the time of the scheduled resection that cannot be stopped for surgery
- Swallowing difficulty that would prevent patient from being able to swallow either temozolomide or Toca FC or severe active mal-absorption

STUDY DESIGN

Arm 1:

Standard of Care temozolomide + RT (Optune allowed)

- Treatment starts 3-6 weeks post surgery

Arm 2:

Standard of Care plus Toca 511, and Toca FC (Optune not allowed)

- Treatment starts 4-6 weeks post surgery

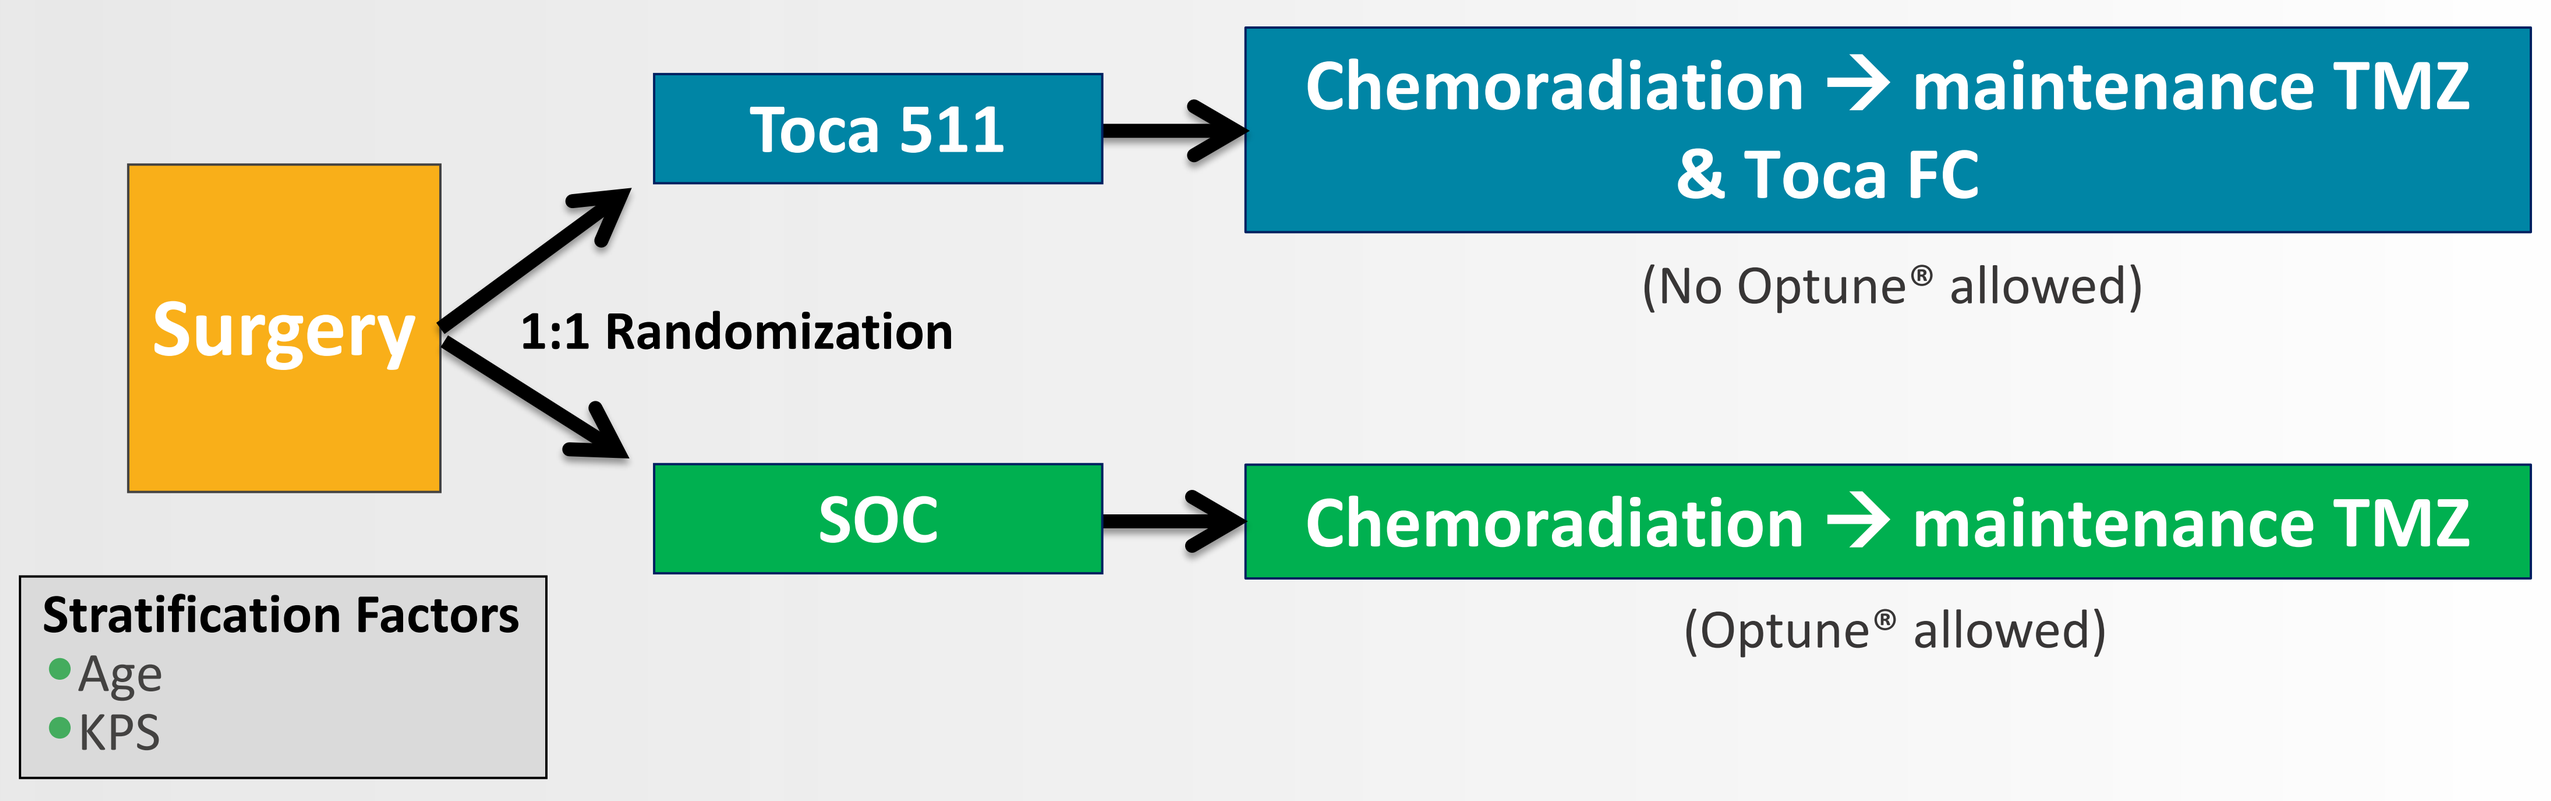
Surgery
1:1 Randomization
Toca 511
Chemoradiation → maintenance TMZ & Toca FC
(No Optune® allowed)
SOC
Chemoradiation → maintenance TMZ
(Optune® allowed)
Stratification Factors
Age
KPS

Figure 3: Treatment Schema

Target Accrual and Activation:

- Target accrual:
 - Phase II: 312 patients
 - Phase III: 900 patients, including Phase II
- Activation: December 2019
- Study Duration: ~ 6 years

If interested in participating in this trial, please contact Erin Moye (MoyeE@nrgoncology.org) for information

NCT Number 04105374